Canada's Lymphedema Magazine

Pathways

Empowering patients and professionals

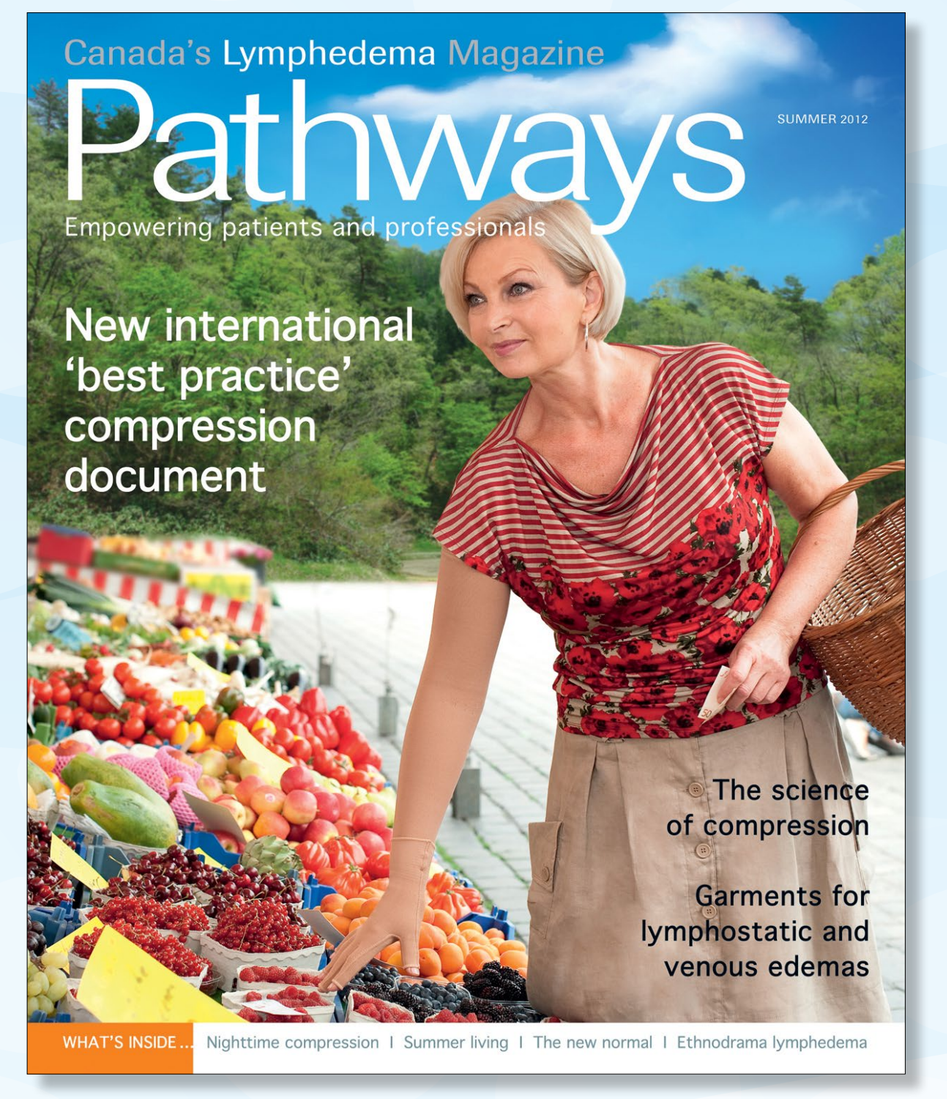
Canada's Lymphedema Magazine
Pathways
Empowering patients and professionals
New international 'best practice' compression document
The science of compression
Garments for lymphostatic and venous edemas

"*Pathways* has been the educational tool that has enabled us to reach out to a wider readership, and to quickly and efficiently share local and world-wide lymphedema news with health professionals, government officials, media and patients across Quebec. The support we receive, being part of this phenomenal extended team across the country, is highly motivational". ***– Rachel Pritzker, President, Lymphedema Association of Quebec***

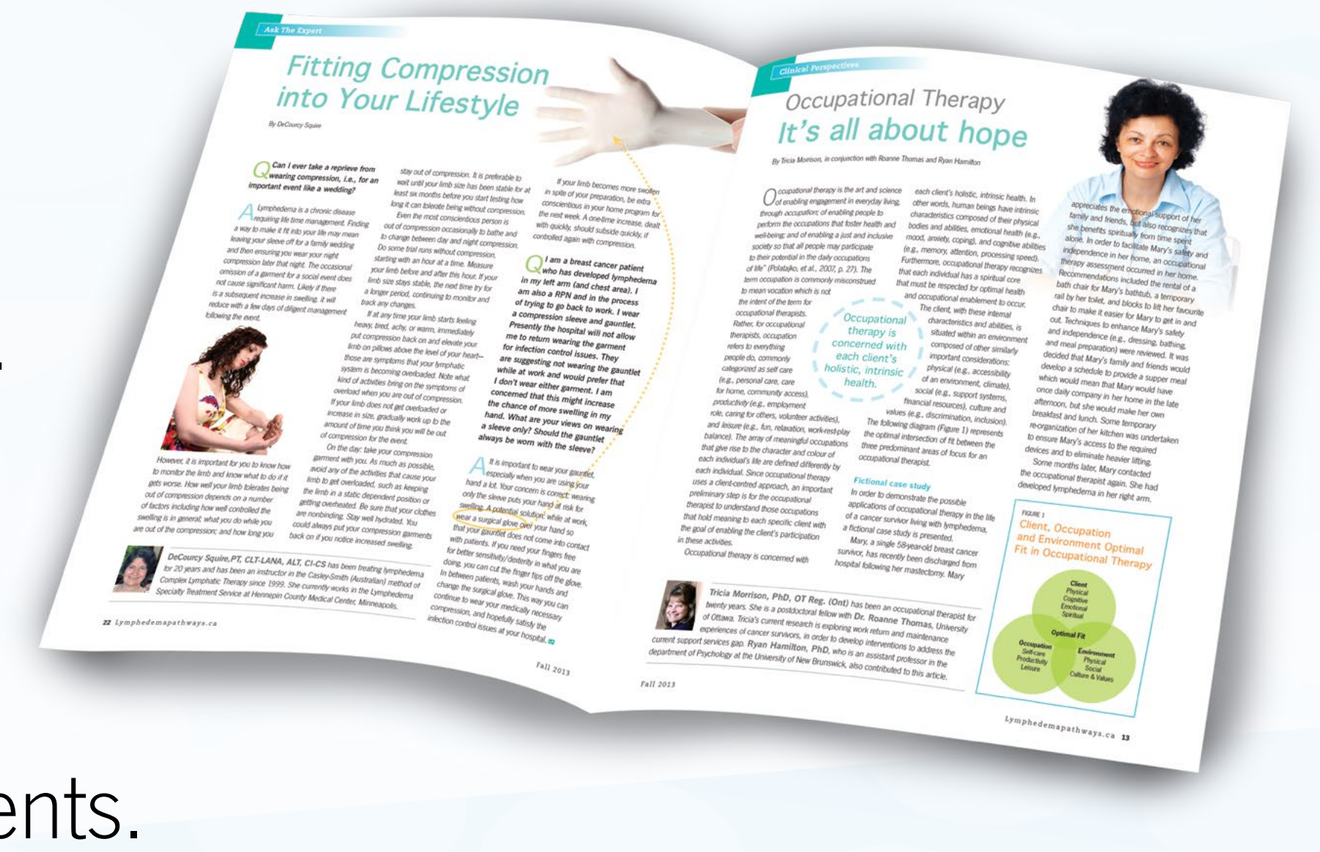
Fitting Compression into Your Lifestyle
Occupational Therapy
It's all about hope

"As a healthcare professional, treating patients with lymphedema, having access to *Pathways* has enabled me to learn from its wonderful information. This translated into improving my practise and positive outcomes for my patients. The magazine helps me explain to patients that they are not alone". ***- Jean Ann Ryan, RNBN CDT, Atlantic Clinical Lymphedema Network***

Congratulations

I offer my congratulations to the Canadian Lymphedema Framework and its provincial affiliates for launching this important tool for education and awareness of lymphedema in Canada.

The CLF has set an important example of how a new lymphedema framework can work with existing lymphedema patient and clinical organizations to unite in one voice for furthering lymphedema care in Canada.

Christine Moffatt, PhD, Chair of the International Lymphoedema Framework

International Lymphoedema Framework — Belong Together

"*Pathways* elevated the visibility of our organization and lent more credibility since we are now seen as part of a larger collaboration. It has increased our ability to help more individuals to lead healthy and hopeful lives with lymphedema. It also created more opportunities for exchanges with other organizations across Canada to share experiences, successes and provide support to each other". ***- Lucette Wesley, President, BC Lymphedema Association***

"With the very first magazine, I already learned something new – so just imagine what all the people who thought they were alone with lymphedema felt when they opened up *Pathways* for the first time. Education and awareness motivates patients to advocate for the help they need". ***- Diane Martin, President, Alberta Lymphedema Association***

"At the time our Canada wide conference calls started, Manitoba did not have an association. Collaborating with other dedicated provincial representatives was the encouragement and support needed to get Manitoba's association off the ground. I am proud to be part of this movement in Canada of creating lymphedema awareness and providing a very professional, evidence-based publication". ***- Kim Avanthay, President, Lymphedema Association of Manitoba***

Canada's Lymphedema Magazine
Pathways
Empowering patients and professionals

Publisher
Pathways is published four times per year by BCS Communications Ltd. on behalf of the Canadian Lymphedema Framework (CLF) and its provincial affiliates.

Editorial Board
David Keast MD FCFP
Winkle Kwan MD FRCPC
Roanne Thomas PhD
Anna Towers MD FCFP
Lucette Wesley CIM
Janice Yurick PT

Editor
Anna Kennedy

Advertising & Submissions
We welcome your ideas, contributions, letters, ad and editorial copy.
All original material is subject to review by the ED Board and must be evidence-based with references.

Subscription Rates
Canada–$25.
International–$50.

Distribution
Western Canada 25%
Central Canada 54%
Eastern Canada 20%
International 1%

lymphedemapathways.ca

The collaboration to launch a unique Canadian Lymphedema Magazine

By Kennedy A, Avanthay K, Martin D, McPherson C, Pritzker R, Ryan J, Tucker-Lloyd E, Wesley L.

Introduction: *Pathways* is a full-colour national magazine providing education for and information to Canadians living with lymphedema, and the various professionals who support their care. The publication is unique in targeting a diverse group of lymphedema stakeholders: patients and caregivers, clinicians, product distributors and fitters, researchers, educators, policy advisors and community organizations.

Context: A 2011 study by the CLF highlighted the need for increased education and awareness of lymphedema. Prior to the creation of the CLF in 2009, eight provincial/regional community organizations were already leading local efforts to educate patients and professionals. There was duplication of efforts.

Objective: To support the CLF's mission to improve the management of lymphedema in Canada and to disseminate lymphedema education and awareness nationally while incorporating customized local content.

Process: Under the leadership of the CLF, monthly cross-Canada calls were initiated. Over two years, eight independent provincial community lymphedema organizations contributed to the concept, design, branding and distribution of the magazine. Sponsoring companies provided financial support.

Content: Canadian and International experts contribute evidence based material for regular columns including Clinical Perspectives, Research Advances, Medical Updates, Case Studies, Patient Perspectives, Ask the Expert, and Resources.

Results: In its first year of publication, *Pathways* surpassed initial targets of readership and industry support. Worldwide response to the magazine has been extremely positive.
Key objectives were met:

- The CLF and its provincial affiliates have united in one strengthened voice.
- The provincial lymphedema associations have a cost-effective tool that can boost their individual membership, attract local advertising revenues and support their provincial advocacy efforts.
- The magazine has provided the CLF with a sustainable revenue source to support its annual operating and project related costs.

Conclusion: The development of *Pathways* provides a model of effective collaboration; several organizations, united in a single passion worked collectively to advance lymphedema education and awareness. Additional collaborative projects are currently under way. LP

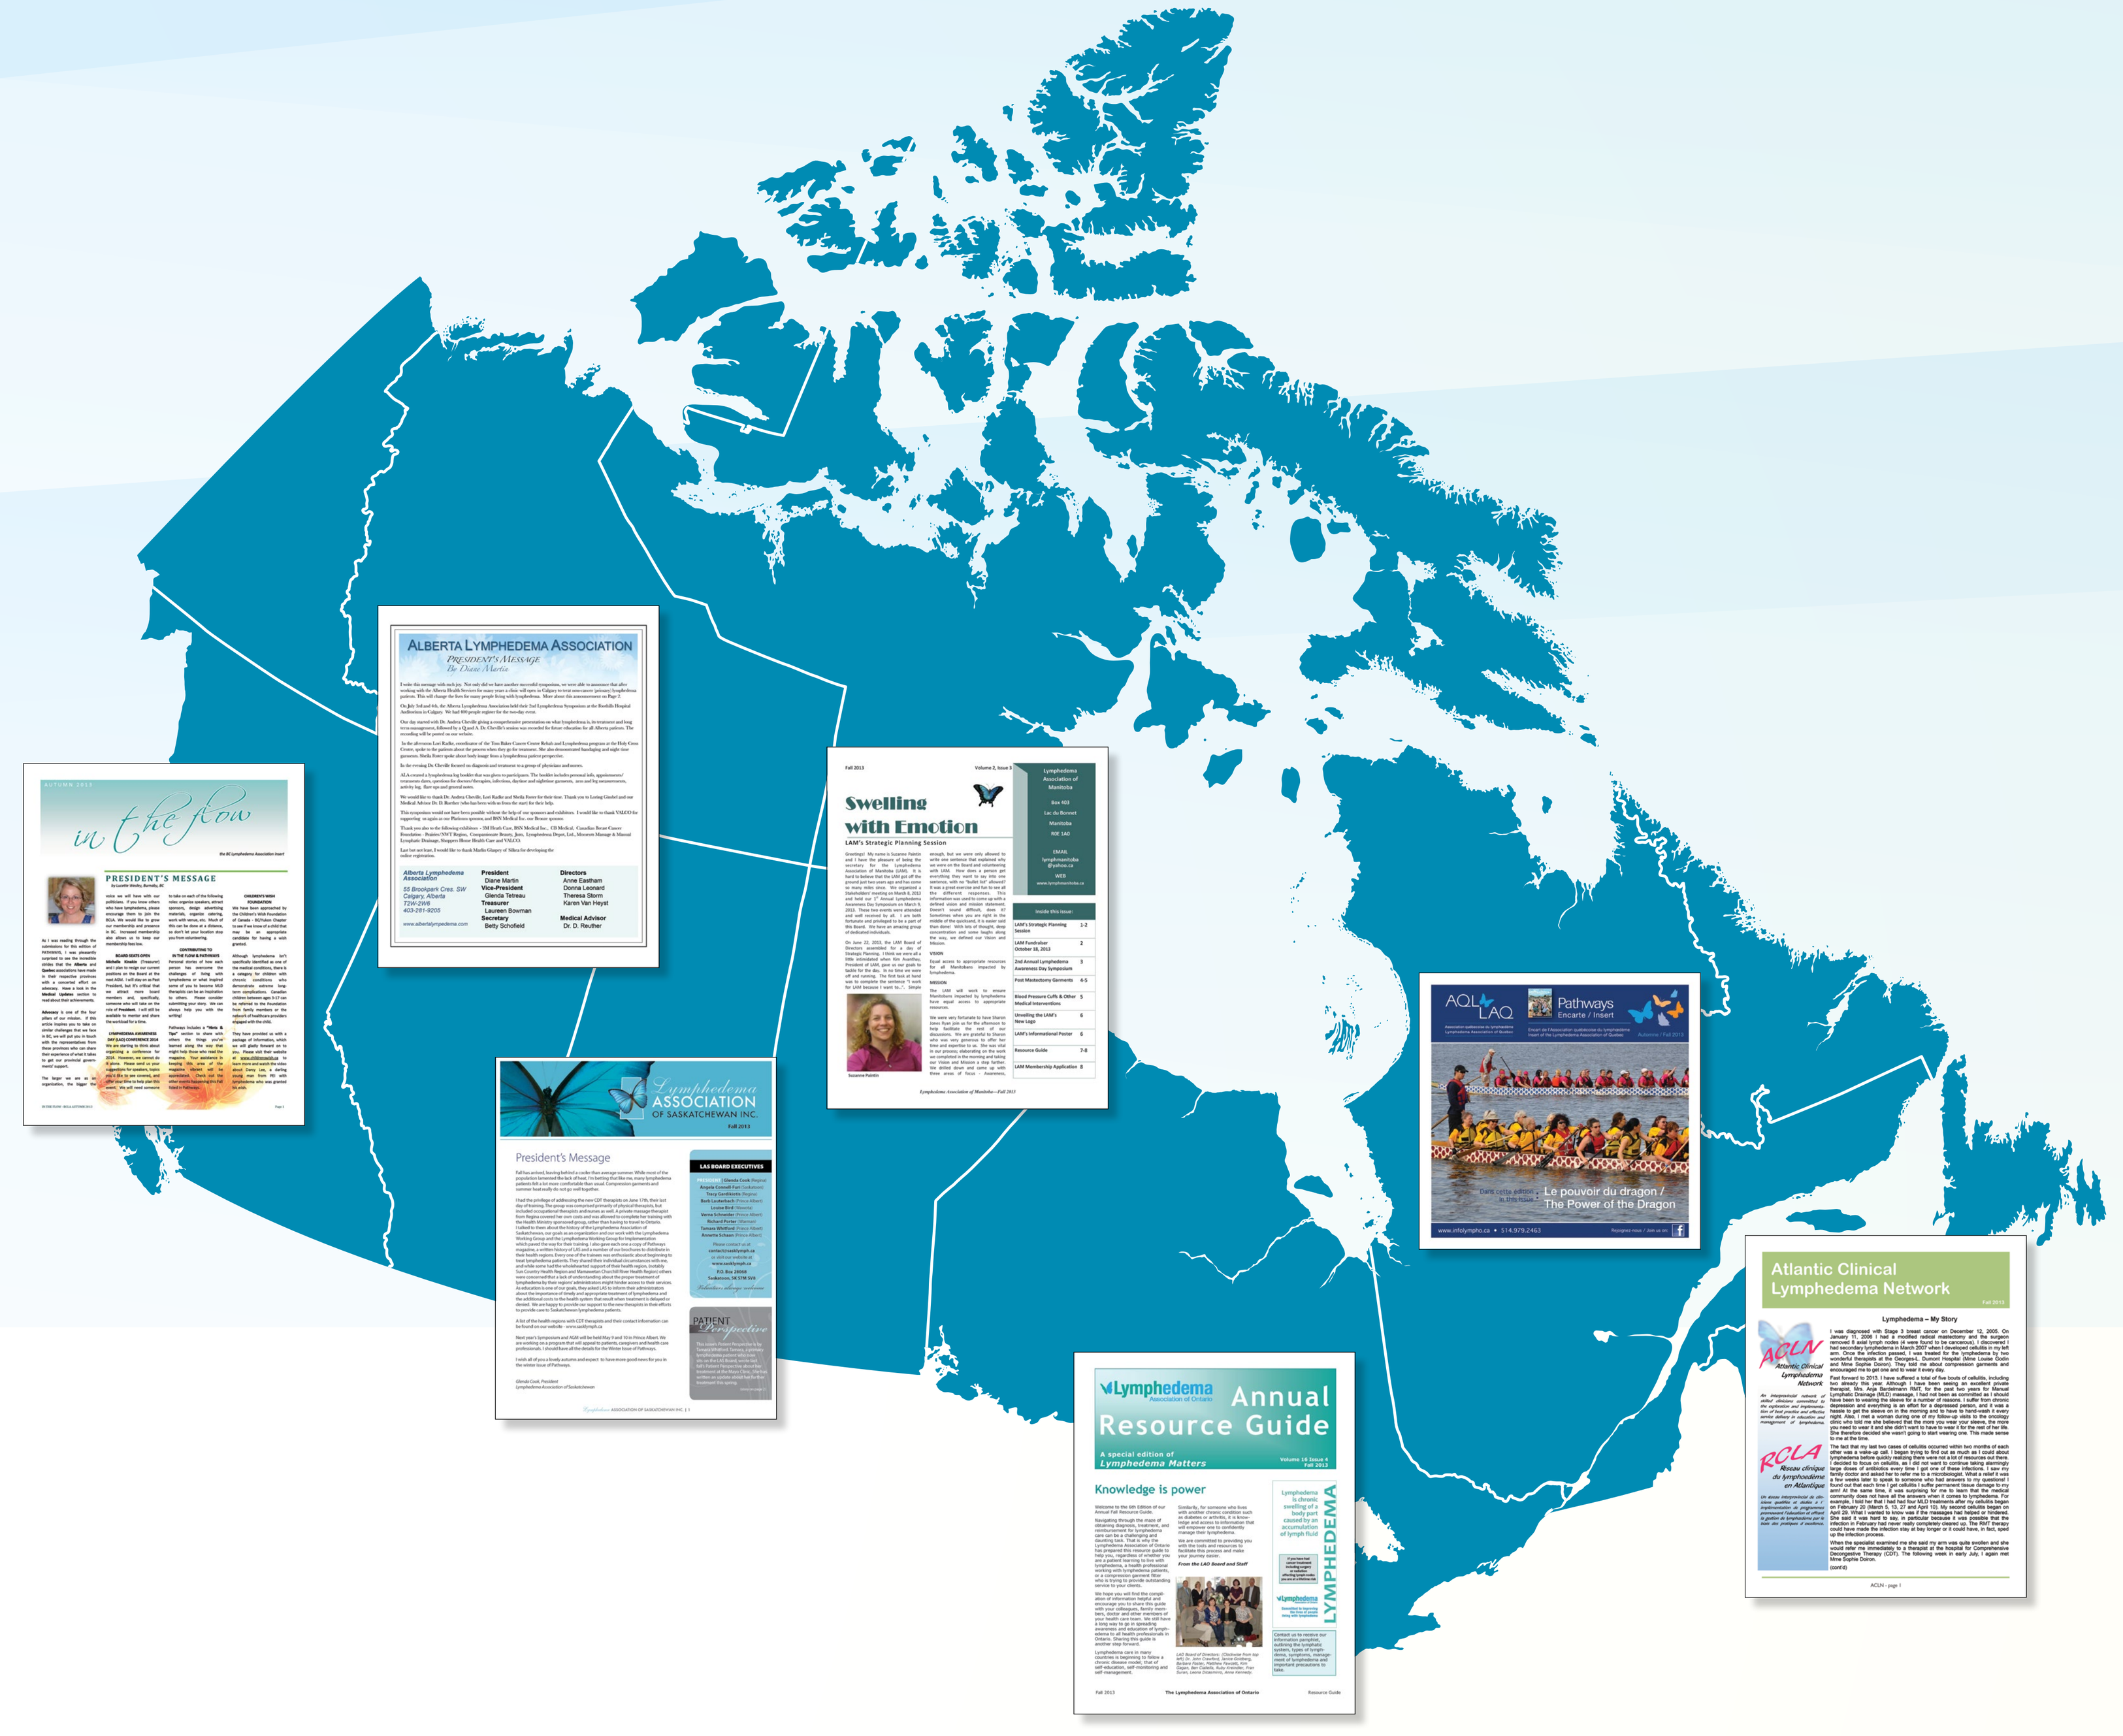
in the flow
Alberta Lymphedema Association
Swelling with Emotion
Lymphedema Association of Saskatchewan Inc.
AQL LAQ
Pathways
Le pouvoir du dragon / The Power of the Dragon
Atlantic Clinical Lymphedema Network
Lymphedema Annual Resource Guide

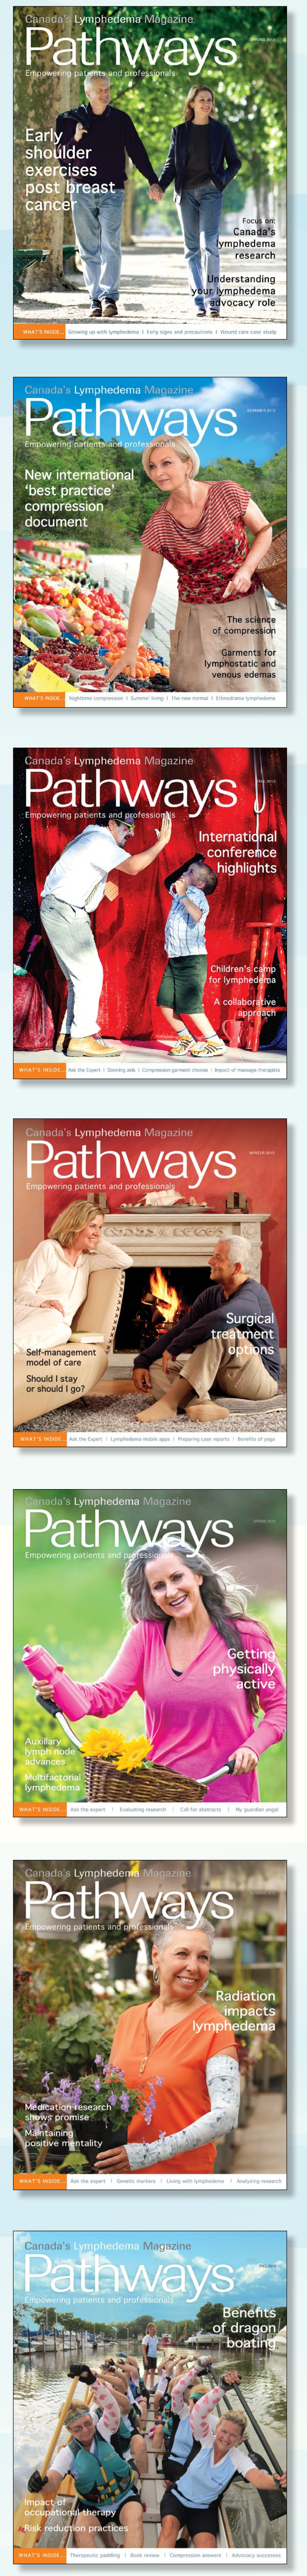

The CLF has affiliate agreements with its provincial partners for the production and distribution of Pathways including customized local inserts.

Alberta Lymphedema Association	www.albertalymphedema.com
Atlantic Clinical Lymphedema Network	www.atlanticlymph.ca
BC Lymphedema Association	www.bclymph.org
Lymphedema Association of **Manitoba**	www.lymphmanitoba.ca
Lymphedema Association of **Ontario**	www.lymphontario.ca
Lymphedema Association of **Quebec**	www.infolympho.ca
Lymphedema Association of **Saskatchewan**	www.sasklymph.ca

About the CLF The Canadian Lymphedema Framework is an academic and patient collaboration, working to improve lymphedema management for lymphedema and related disorders in Canada.

Charitable Registration 85160 1260 RR0001

The Canadian Lymphedema Framework (CLF) wishes to thank key industry partners for their financial support in making this magazine a reality.

Platinum Sponsorship BSN medical, medi

Gold Sponsorship 3M

Silver Sponsorship PASCOE Natural Medicine, Juzo Since 1912